A Phase 3 Trial of Firdapse™ Tablets in Lambert-Eaton Myasthenic Syndrome

Charles W Gorodetzky, MD, PhD,[1] Mazen M Dimachkie, MD,[2] M Douglas Winship, BS,[1] Shin J Oh, MD,[3] and the LEMS Trial Study Team

[1]Catalyst Pharmaceutical Partners, Inc., Coral Gables, FL, United States, 33134; [2]University of Kansas Medical Center, Kansas City, KS, United States, 66160; [3]University of Alabama-Birmingham, Birmingham, AL, United States, 35233

INTRODUCTION

Lambert-Eaton Myasthenic Syndrome (LEMS) is a rare autoimmune disease with a prevalence of ~3,000 patients in the United States and Canada. The primary symptom is muscle weakness—the result of autoantibodies to voltage-gated calcium channels, causing a reduction in the amount of acetylcholine (ACh) released from nerve terminals (Figure 1).

The most common clinical presentations of LEMS are proximal muscle weakness and easy fatigability that may lead to difficulty walking and climbing stairs. The classical triad of LEMS includes proximal leg weakness, hyporeflexia or areflexia, and cholinergic dysautonomia (dry mouth, impotence, etc.) Ocular bulbar weakness is rare. Symptoms can be life threatening when the weakness involves respiratory muscles. Approximately 50% of LEMS patients have small cell lung cancer.

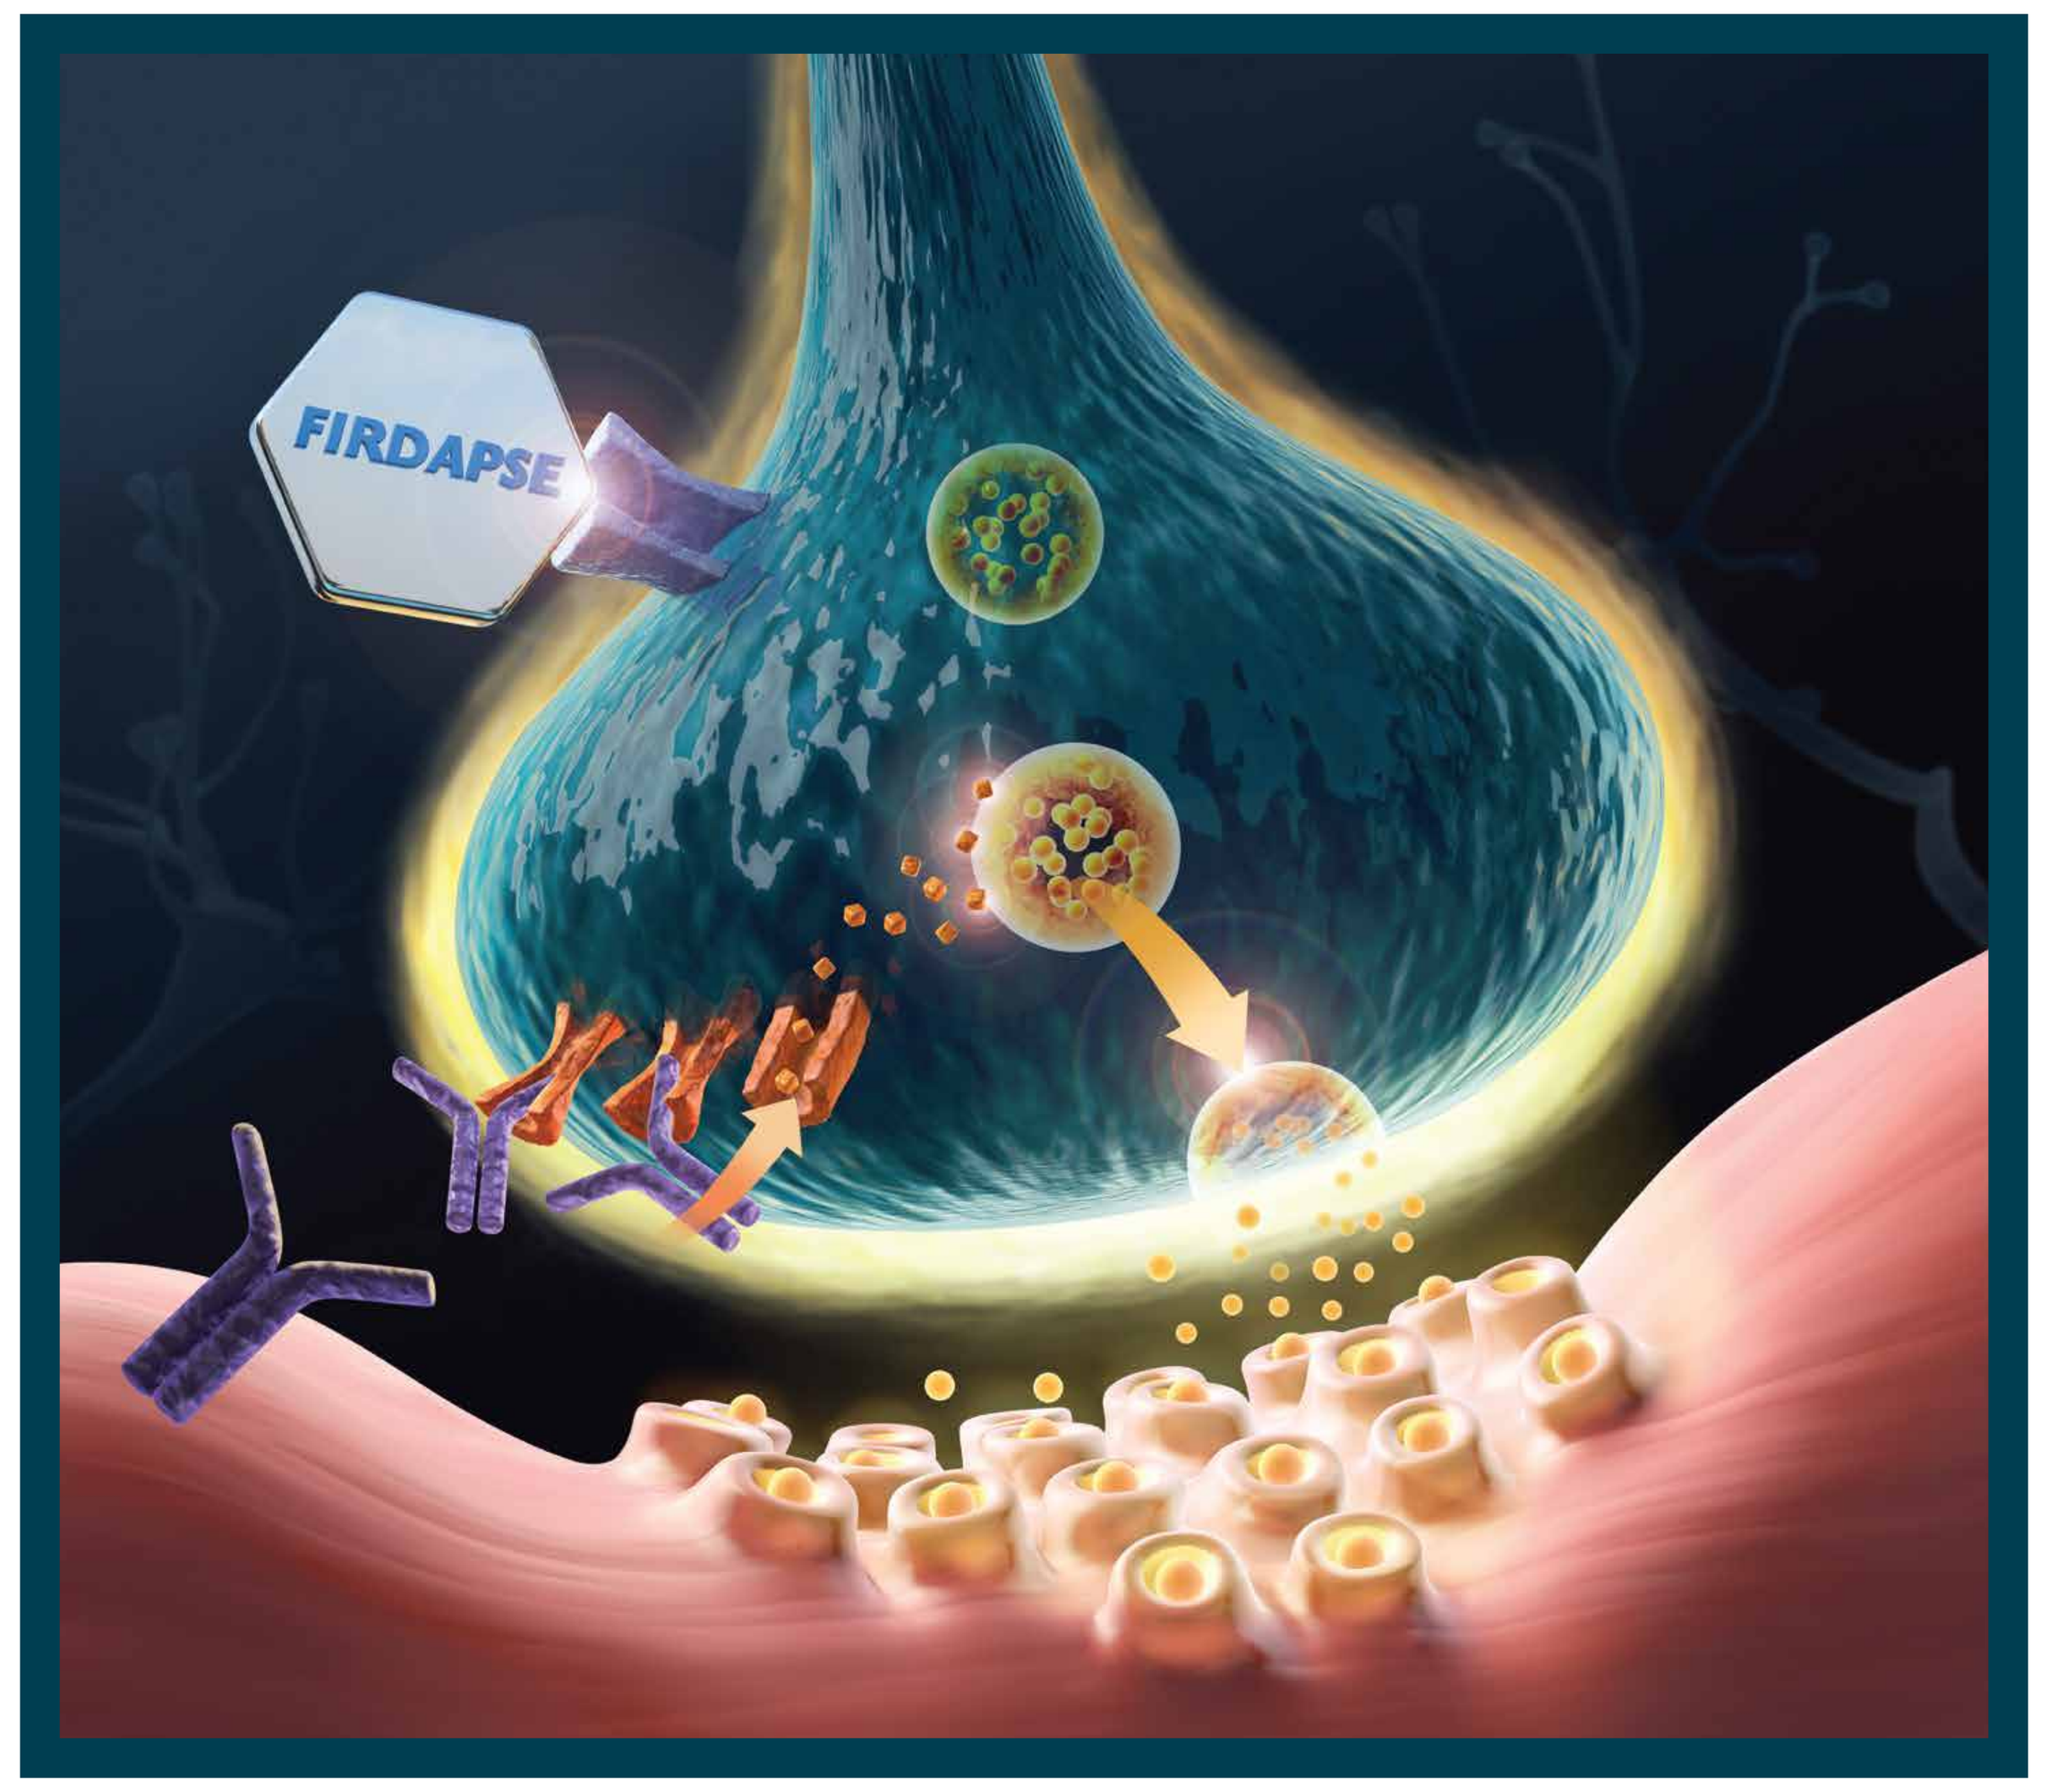
FIRDAPSE

Figure 1. Mechanism of action of Firdapse™

Mechanism of Action of Amifampridine

Nonspecific voltage-dependent potassium (K+) channel blocker

Causes depolarization of the presynaptic membrane and slows or inhibits repolarization

Results in opening of slow voltage-dependent calcium (Ca^{2+}) channels and allows a subsequent influx of Ca^{2+}

Induces exocytosis of the synaptic vesicles containing ACh, into the synaptic cleft

Enhances neuromuscular transmission, providing improved muscle function

OBJECTIVES

To evaluate the efficacy and safety, including the long-term safety, of amifampridine (3,4-DAP) phosphate as a symptomatic treatment for patients with LEMS, in a Phase 3, multicenter, double-blind, placebo-controlled, randomized discontinuation study followed by an open-label extension period.

STUDY RATIONALE

Currently, there is no FDA-approved treatment for LEMS in the United States. The following represent the two main approaches to managing patients.

1. Symptomatic treatment (eg, pyridostigmine, amifampridine, guanidine)
2. Immunologic treatments (eg, intravenous immunoglobulin (IVIG), plasma exchange, immunoadsorption, prednisone, azathioprine), aimed at suppressing the autoimmune reaction
3. Cancer treatment is critical, if cancer is found

Amifampridine formulations that are available on a compassionate-use basis have uncertain pharmaceutical quality. The salt species of 3,4-DAP has been shown to have superior stability compared with base.[1]

To overcome the stability and quality issues present in the compounded base formulation, a phosphate salt formulation of amifampridine (3,4-diaminopyridine phosphate) was developed.

This formulation received marketing approval by the European Commission, as Firdapse™, for the symptomatic treatment of patients with LEMS, in December 2009.

OVERVIEW OF STUDY DESIGN

Randomization
38 patients randomized, 1:1

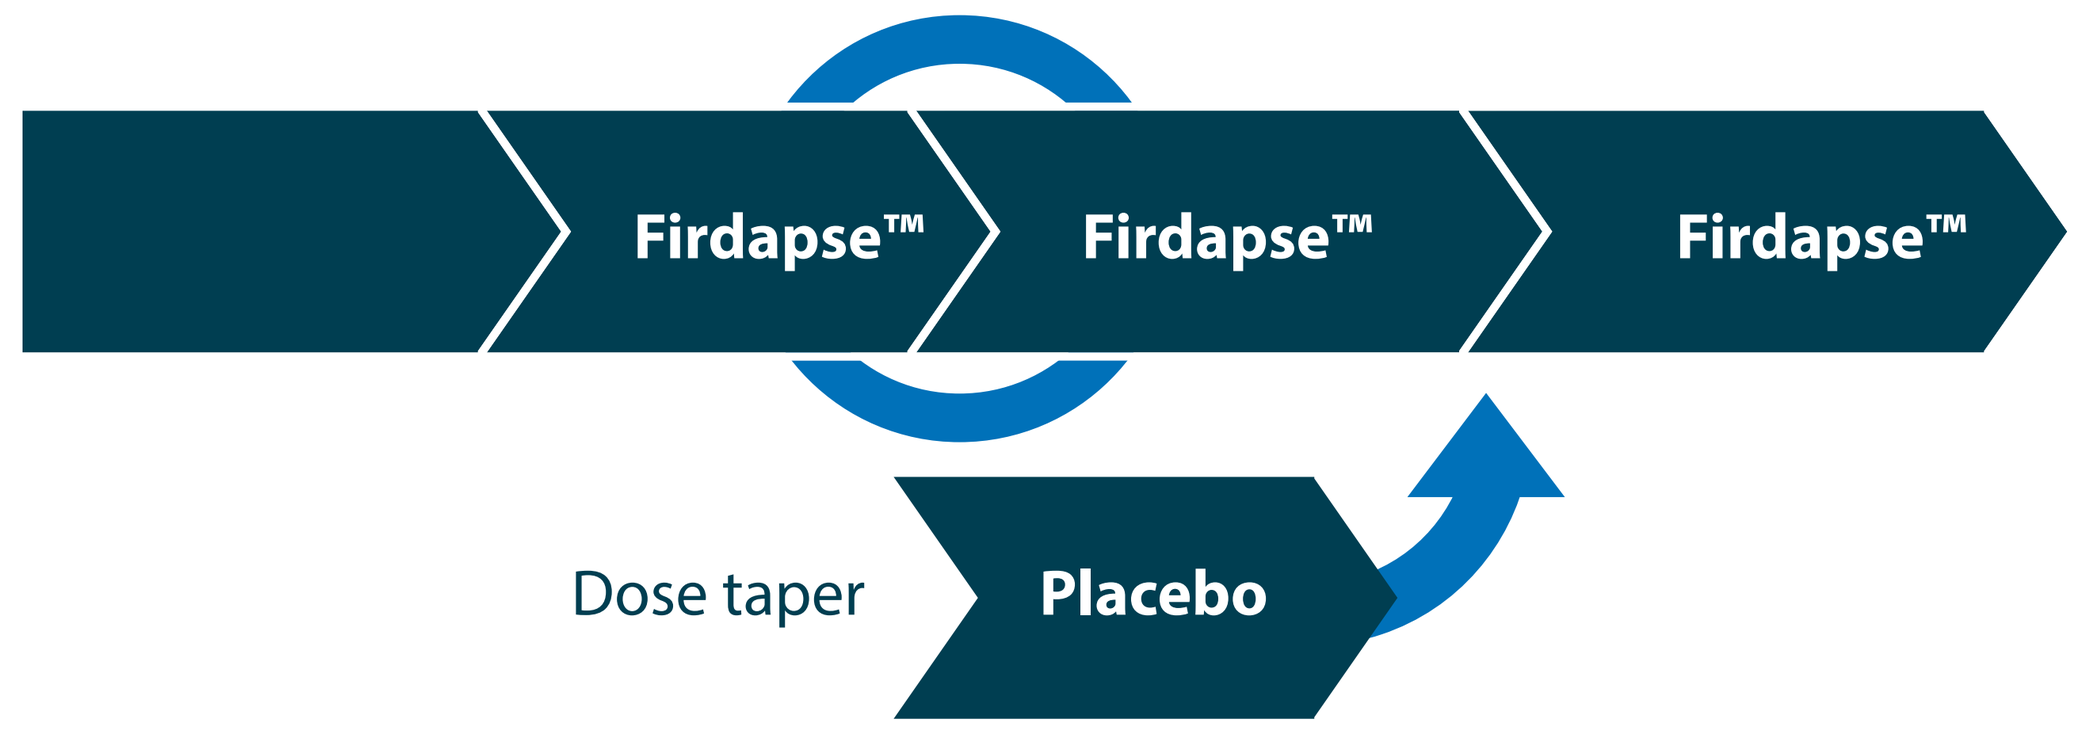
Firdapse™
Firdapse™
Firdapse™
Dose taper
Placebo

Screening	Open-Label Run-In	Double-Blind Treatment Phase		Open-Label Safety Extension
Days 1-4	Days 7-91	Days 1-7	Days 8-14	Up to 2 years
Efficacy/Baseline assessments screening	Efficacy/Eligibility assessments and dose adjustments	Efficacy assessments on Day 1	Efficacy assessments on Days 8 and 14	Safety assessments and dose adjustments
		1. Quantitative Myasthenia Gravis (QMG); Subject Global Impression (SGI) 2. Clinical Global Impression of Improvement (CGI-I); Timed 25-Foot Walk (T25FW) 3. Compound Muscle Action Potential (CMAP)		Not required for New Drug Application (NDA) filing and approval

Figure 2. Study design summary

DETAILS OF STUDY DESIGN

Part 1 Open-Label Run-In (Days 7 to 91)—54 Subjects

Baseline QMG* score for 38 patients with LEMS → ≥91 days of 3,4-DAP (30-80 mg/day) taken in divided doses of 10 or 20 mg tid or qid → 7-day stable period

QMG—Day 30 | QMG—Day 60 | QMG—Day 90

*To be eligible for Part 2, QMG score must improve by ≥3 points from baseline.

Part 2 Double-Blind Treatment Discontinuation. Randomization (7 days)—38 Subjects

GROUP A: ~50% maintained on amifampridine

GROUP B: ~50% tapered off amifampridine over a 7-day period, followed by 7 days on placebo

Efficacy Assessment

Primary
- Quantitative examination of muscle strength measured by QMG score
- SGI score

Secondary
- Investigator-perceived CGI-I score
- T25FW results

Part 3 Double-Blind Treatment (7 days)—38 Subjects

GROUP A: Continue on amifampridine for 7 more days

GROUP B: Continue on placebo for 7 more days

Efficacy and safety assessments performed on Day 8 and Day 14

Part 4 Open-Label Extension—40 Subjects

1. Patients who completed Parts 1, 2, and 3
2. Patients who received, per protocol, rescue treatment with amifampridine phosphate
3. Patients who participated in Part 1 but did not establish 7 days of stable amifampridine phosphate dosing when the 36th patient was randomized in Part 2

- Amifampridine dosing will began on Day 15 following randomization. Patients were seen on Day 19 (± 2 days), then monthly for the next 2 visits, and then quarterly, thereafter
- Safety assessments performed at each visit

STUDY PLAN

~36 patients completed Part 3 of the study

Diagnosis and Criteria for Inclusion

- ≥18 years of age
- Confirmed diagnosis of LEMS, as documented by acquired (typical) proximal muscle weakness, and at least 1 of the following:
 - Repetitive nerve stimulation test findings (CMAP that increases at least 2-fold after maximum voluntary contraction of the tested muscle)
 - Positive anti-P/Q type voltage-gated calcium-channel antibody test
- A QMG score of ≥5 for patients with no prior symptomatic treatment for LEMS
- If receiving treatment for LEMS, presentation of some signs and/or symptoms consistent with LEMS
- If receiving treatment with amifampridine for LEMS, a normal respiratory function as defined by a forced vital capacity (FVC) ≥80% of predicted
- If not receiving treatment with amifampridine for LEMS and no history of other current respiratory disease, an FVC ≥60% of predicted
- Normal swallowing function as defined by the ability to swallow 4 ounces of water without coughing or throat clearing (score of 0 on this dimension of QMG)
- If receiving peripherally acting cholinesterase inhibitors (eg, pyridostigmine), a stable dose of cholinesterase inhibitors is required for at least 7 days prior to screening
- If receiving permitted oral immunosuppressants (eg, prednisone or other corticosteroid, azathioprine, mycophenolate), a stable dose is required for at least 90 days prior to screening

EFFICACY RESULTS

Statistical analyses using a mixed-effect model of repeated measures (MMRM) demonstrated that patients who received placebo had a greater worsening in QMG and SGI scores than did patients who received amifampidine.

Change in QMG scores from baseline (Day 1, Part 2) to Day 14 (Part 3): amifampridine vs placebo

In the randomization phase of the trial, the change in QMG scores from baseline to Day 14, one of the primary endpoints, reached statistical significance, with the least square (LS) mean for QMG score increasing by 2.2 in placebo-treated patients, and increasing by 0.4 in amifampridine-treated patients (Figure 3; P=0.0452). In addition, the average change in QMG score of 2.4 points for those patients who were discontinued from the test medication was larger than the 2-point minimum change generally considered by medical experts to be clinically significant.[2]

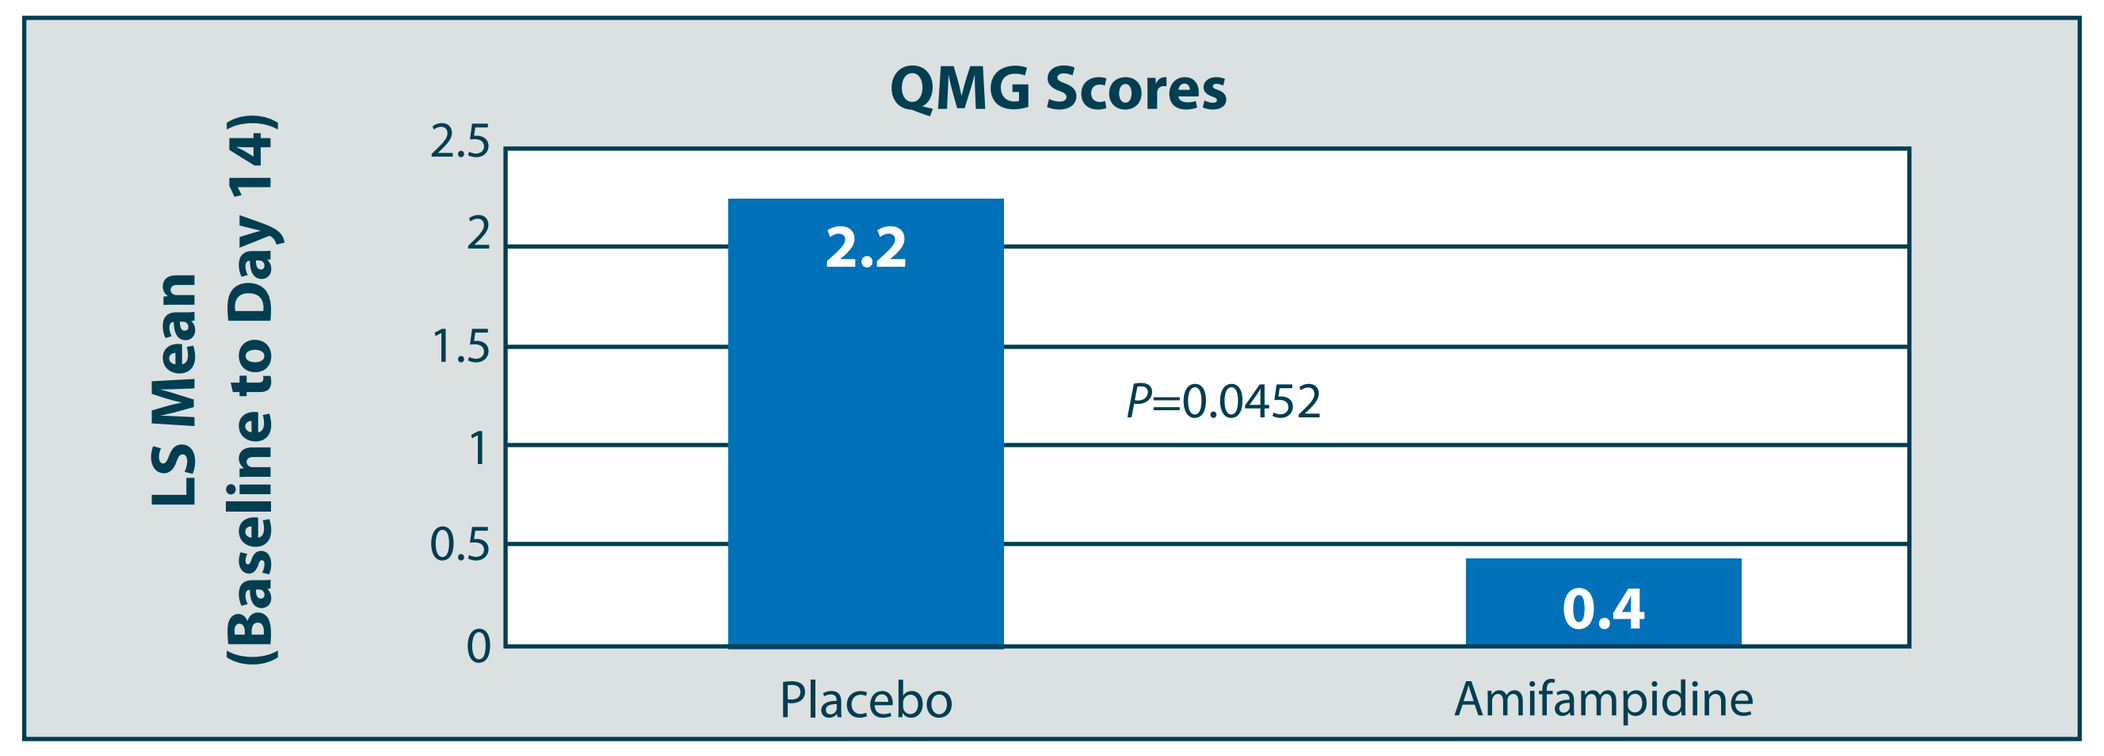
QMG Scores
LS Mean (Baseline to Day 14)
2.5
2
1.5
1
0.5
0
2.2
0.4
P=0.0452
Placebo
Amifampidine

Figure 3. Results from MMRM for the change in QMG scores from baseline (Day 1, Part 2) to Day 14 (end of Part 3)

EFFICACY RESULTS (CONTINUED)

Change in SGI scores from baseline (Day 1, Part 2) to Day 14 (Part 3): amifampridine vs placebo

Patients who were receiving amifampridine (Firdapse™) tablets at the baseline time point (Day 1) reported, on average, that they were "pleased" (SGI score of 5.9 ± 1.2) with the test medication while they were receiving it. Upon being switched to placebo tablets, their opinions dropped, on average, 2.7 ± 2.3 points. The LS mean was 2.6 for the placebo group and -0.8 for the amifampridine group, a difference of 1.8 ± 0.6 (Figure 4; P=0.0028), corresponding to a patient assessment, of "mixed" for the placebo tablets. This substantial change in patients' assessments, to a worsening of their condition while receiving placebo, is considered clinically significant.[3]

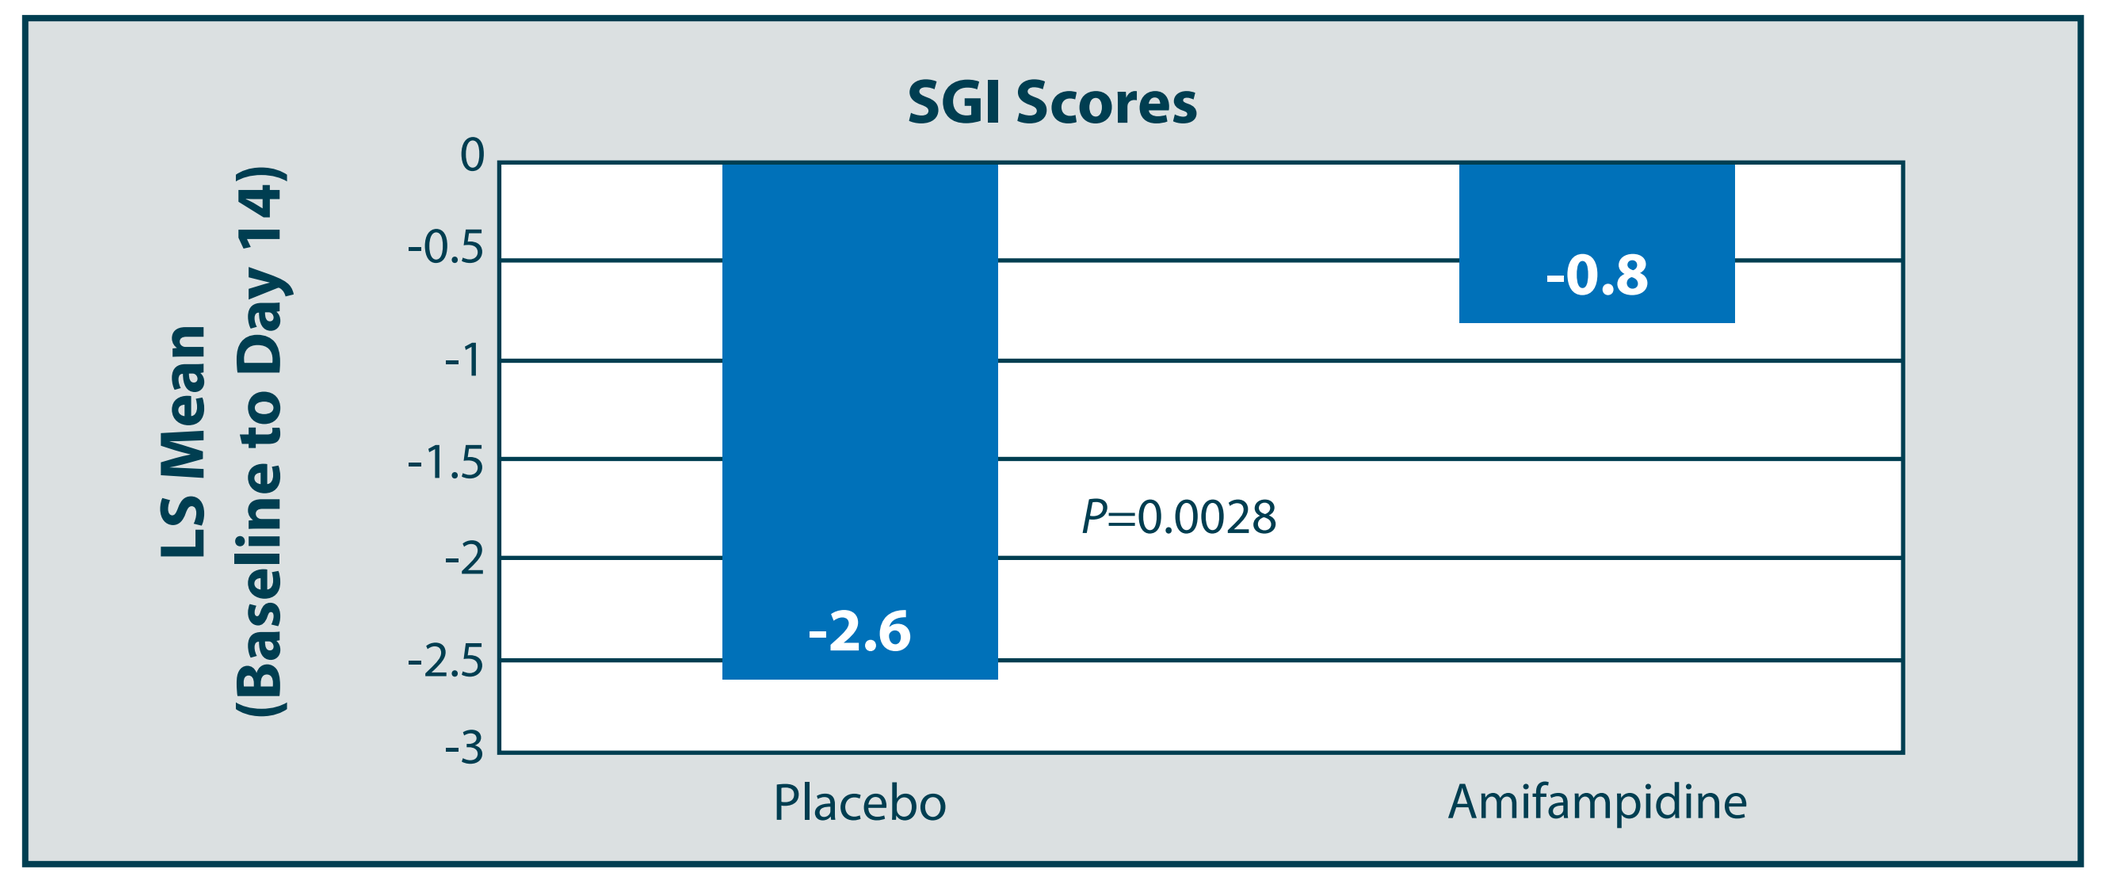
SGI Scores
LS Mean (Baseline to Day 14)
0
-0.5
-1
-1.5
-2
-2.5
-3
-2.6
-0.8
P=0.0028
Placebo
Amifampidine

Figure 4. Results from MMRW for the change in SGI scores from baseline (Day 1, Part 2) to Day 14 (end of Part 3)

SECONDARY ENDPOINTS

The change in investigator-perceived CGI-I score, indicating a of worsening of disease symptoms during the 2-week, double-blind testing period, also reached statistical significance (Table 1).

The CGI-I score at Day 14 for placebo-treated patients was 4.7; (P=0.0267). The other secondary endpoint, a change in T25FW speed at Day 14, showed a quantitative worsening of 9.67 feet/min in the placebo-treated group (Table 1). However, the magnitude of the change, relative to the variance inherent in this test, did not reach statistical significance for this endpoint , due to the small sample size.

Table 1. Secondary Endpoint Results

Endpoint	Clinical Outcome	Significance
CGI-I	Placebo subjects' score at Day 14 was 4.7	P=0.0267
T25FW	Placebo subjects' speed deceased 9.67 feet/min	P>0.05 (NS)

SAFETY AND TOLERABILITY

- Currently, 40 subjects are participating in the 2-year follow-up phase of the study
 - 7 patients continue treatment under an extension protocol
 - 1 patient reportedly was switched to a commercial product available in Germany
- No serious adverse events attributable to the drug have occurred to date. Amifampridine (Firdapse™) tablets were generally safe and well tolerated; side effects were benign, consisting of perioral and digital paresthesia, and infections (Table 2).

SAFETY TABLE

Table 2. Treatment-Related Adverse Events (TEAEs)

Part 1. Summary TEAEs During Open-Label Run-In (>20%)		
	Overall	
Percentage of patients with TEAEs	69.8%	
Gastrointestinal disorders	52.8%	
Nervous system disorders	45.3%	
Infections and infestations	20.8%	
Part 2. Summary TEAEs During Double-Blind Treatment Discontinuation		
	Placebo	**Amifampridine**
Percentage of patients with TEAEs	12.6%	37.5%
Nervous system disorders	0%	12.5%
Infections and infestations	4.5%	12.5%
Part 3. Summary TEAEs During the Double-Blind Treatment Phase		
	Placebo	**Amifampridine**
Percentage of patients with TEAEs	27.1%	18.8%
Infections and infestations	4.5%	18.8%

CONCLUSIONS

- This Phase 3 trial in 38 patients with LEMS demonstrated the safety and efficacy of amifampridine (Firdapse™) and that it was well tolerated
- Amifampridine met its coprimary endpoints of improving physician-rated QMG score (P=0.0452) and patient-reported satisfaction in the SGI score (P=0.0028) compared with placebo-treated patients
- A secondary endpoint, investigator-perceived CGI-I score, was also statistically significant (P=0.0267)
- The secondary endpoint of T25FW speed showed a difference between amifampridine and placebo that was not statistically significant, due to the small size of the patient population in the study and the inherent variability in the test

2014 Annual Meeting of the American Neurological Association
Baltimore Marriott Waterfront, Baltimore, MD
Poster No. S737WIP October 12 – 14, 2014

References: 1. Raust JA, Goulay-Dufaÿ, Le Hoang MD, Pradeau D, Guyon F, Do B. Stability studies of ionised and non-ionised 3,4-diaminopyridine: hypothesis of degredation pathways and chemical structure of degradation products. *J Pharm Biomed Anal.* 2007;43(1):83-88. **2.** Sanders DB, Massey JM, Sanders LL, Edwards LJ. A randomized trial of 3,4-diaminopyridine in Lambert-Eaton myasthenic syndrome. *Neurology.* 2000;54(3):603-607. **3.** Goodman AD, Brown TR, Krupp LB, Schapiro RT, et al. Sustained-release oral fampridine in multiple sclerosis: a randomised, double-blind, controlled trial. *Lancet.* 2009;373(9665):732-738.

S737WIP

© 2014 Catalyst Pharmaceutical Partners, Inc. (Catalyst Pharmaceuticals) CPP-14005